PP01.35

Serplulimab vs atezolizumab added to chemotherapy in patients with treatment-naive ES-SCLC in the United States – ASTRIDE trial

Afshin Dowlati, MD[1]; Evan Wu, MD, PhD[2]; Sonam Puri, MD[3]; Omar Abughanimeh, MBBS[4]; Jing Li[5]; David Gandara, MD[6]
[1]University Hospitals, Case Western Reserve University, Cleveland, OH; [2]University of Hawaii Cancer Center/Hawaii Pacific Health, Honolulu, HI; [3]Huntsman Cancer Institute - University of Utah Health, Salt Lake City, UT; [4]University of Nebraska Medical Center, Omaha, NE; [5]Shanghai Henlius Biotechnology Co., Ltd; [6]University of California, Davis, CA

Henlius

FOSUN PHARMA USA INC.

Background

- Small cell lung cancer (SCLC) is the most aggressive subtype of lung cancer, accounting for ~15% of all lung cancer cases[1]
- SCLC is classified as either limited stage small cell lung cancer (LS-SCLC) or extensive stage small cell lung cancer (ES-SCLC)[1]
 - Approximately 2 out of 3 patients with SCLC have ES-SCLC when diagnosed[1]
- Following approvals of atezolizumab (2019) and durvalumab (2020) combination therapies based on the Phase 3 IMpower133 and CASPIAN studies, respectively, anti–PD-L1 + etoposide-platinum chemotherapy became the SOC in ES-SCLC; these therapies have offered ~2 month OS benefit compared to chemotherapy alone[2-4]
- Due to significant unmet medical need for SCLC, there are multiple ongoing studies of serplulimab, exploring its safety and efficacy in patients with both LS-SCLC and ES-SCLC[5]
- Results from the ASTRUM-005 study, a global, randomized, placebo-controlled Phase 3 clinical study, led to the approval of serplulimab for the 1L treatment of ES-SCLC in China, making it the world's first anti–PD-1 monoclonal antibody (mAb) to be approved in SCLC[6,7]
 - Serplulimab + chemotherapy showed an improvement in median OS by 4.7 months vs placebo + chemotherapy (15.8 months vs 11.1 months; HR 0.62 [95% CI: 0.50-0.76]; *P*<0.001) in treatment-naive patients with ES-SCLC[8]
- Fosun Pharma entered into an exclusive license agreement with Shanghai Henlius Biotech, Inc. for the commercialization of serplulimab in the US[9]
- ASTRIDE, a head-to-head, US-based study of serplulimab + chemotherapy vs atezolizumab + chemotherapy for the treatment of 1L ES-SCLC, aims to confirm the ASTRUM-005 findings in a US-based ES-SCLC population[10]

Mechanism of action of serplulimab

- Serplulimab is a novel anti–PD-1 mAb with a unique mode of recognition of the PD-1 receptor compared with currently available anti–PD-1 mAbs[11]
 - While serplulimab shows greater overlap of its epitope region with pembrolizumab than nivolumab, it has an opposite heavy chain and light chain usage vs pembrolizumab, resulting in a different spatial interaction with PD-1[11,12]
- In vitro, serplulimab was able to specifically bind the PD-1 receptor and efficiently block the PD-L1 and PD-L2 signaling pathway with relatively higher potency than nivolumab[11]
- In vivo, serplulimab demonstrated similar antitumor activity as pembrolizumab and the potential for complete tumor eradication, while nivolumab only demonstrated tumor growth inhibition[11]

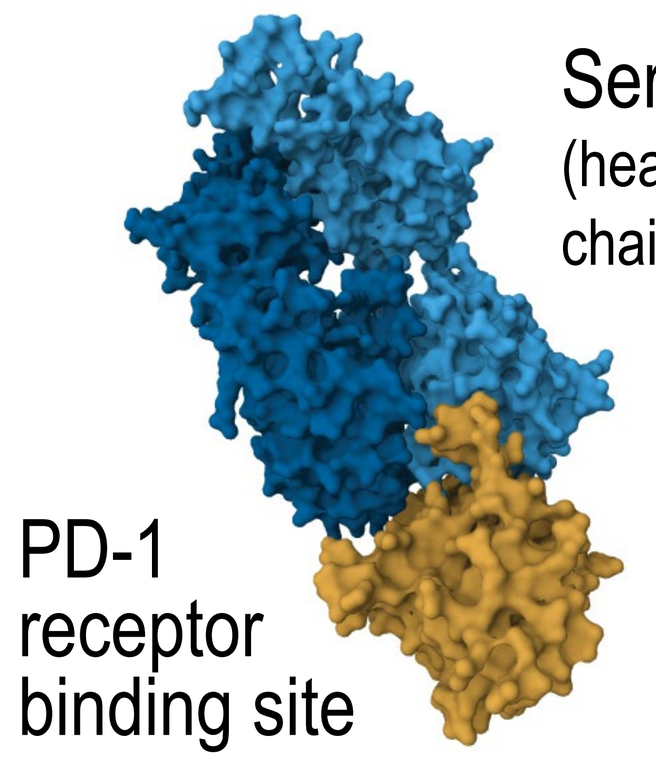
Serplulimab
(heavy and light chains)
PD-1 receptor binding site

Study design: Phase 3, randomized, open-label study

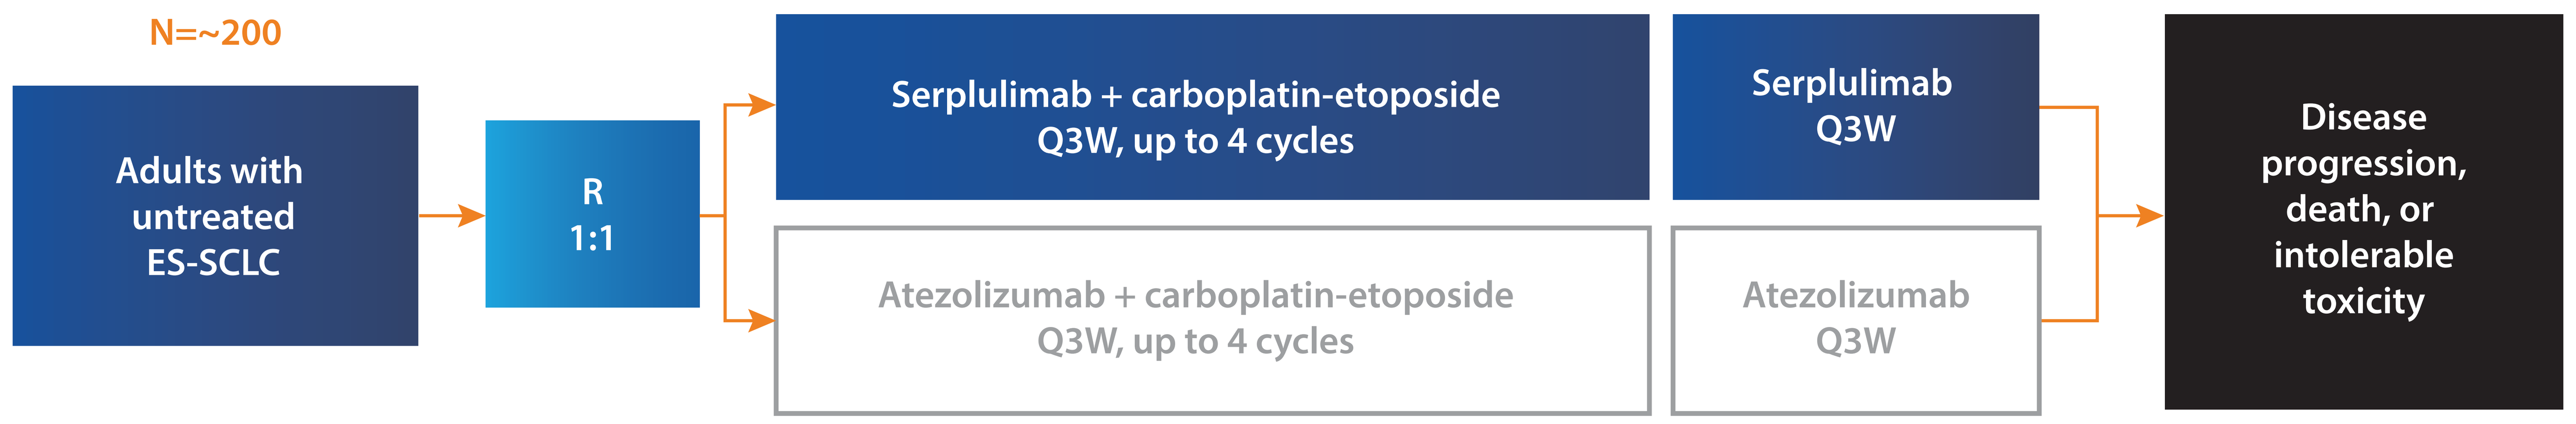
N=~200
Adults with untreated ES-SCLC
R 1:1
Serplulimab + carboplatin-etoposide Q3W, up to 4 cycles
Serplulimab Q3W
Atezolizumab + carboplatin-etoposide Q3W, up to 4 cycles
Atezolizumab Q3W
Disease progression, death, or intolerable toxicity

Key endpoints

Primary: OS

Secondary: PFS, ORR, DOR, QoL, PK/PD, correlative biomarker analyses

Safety: AEs

Study status

- ASTRIDE is currently recruiting participants; an estimated 200 participants from 90 sites across the US will be enrolled
- This is an ongoing study; enrollment began on November 18, 2022
- The study has an estimated primary completion date of June 2024

ASTRIDE sites

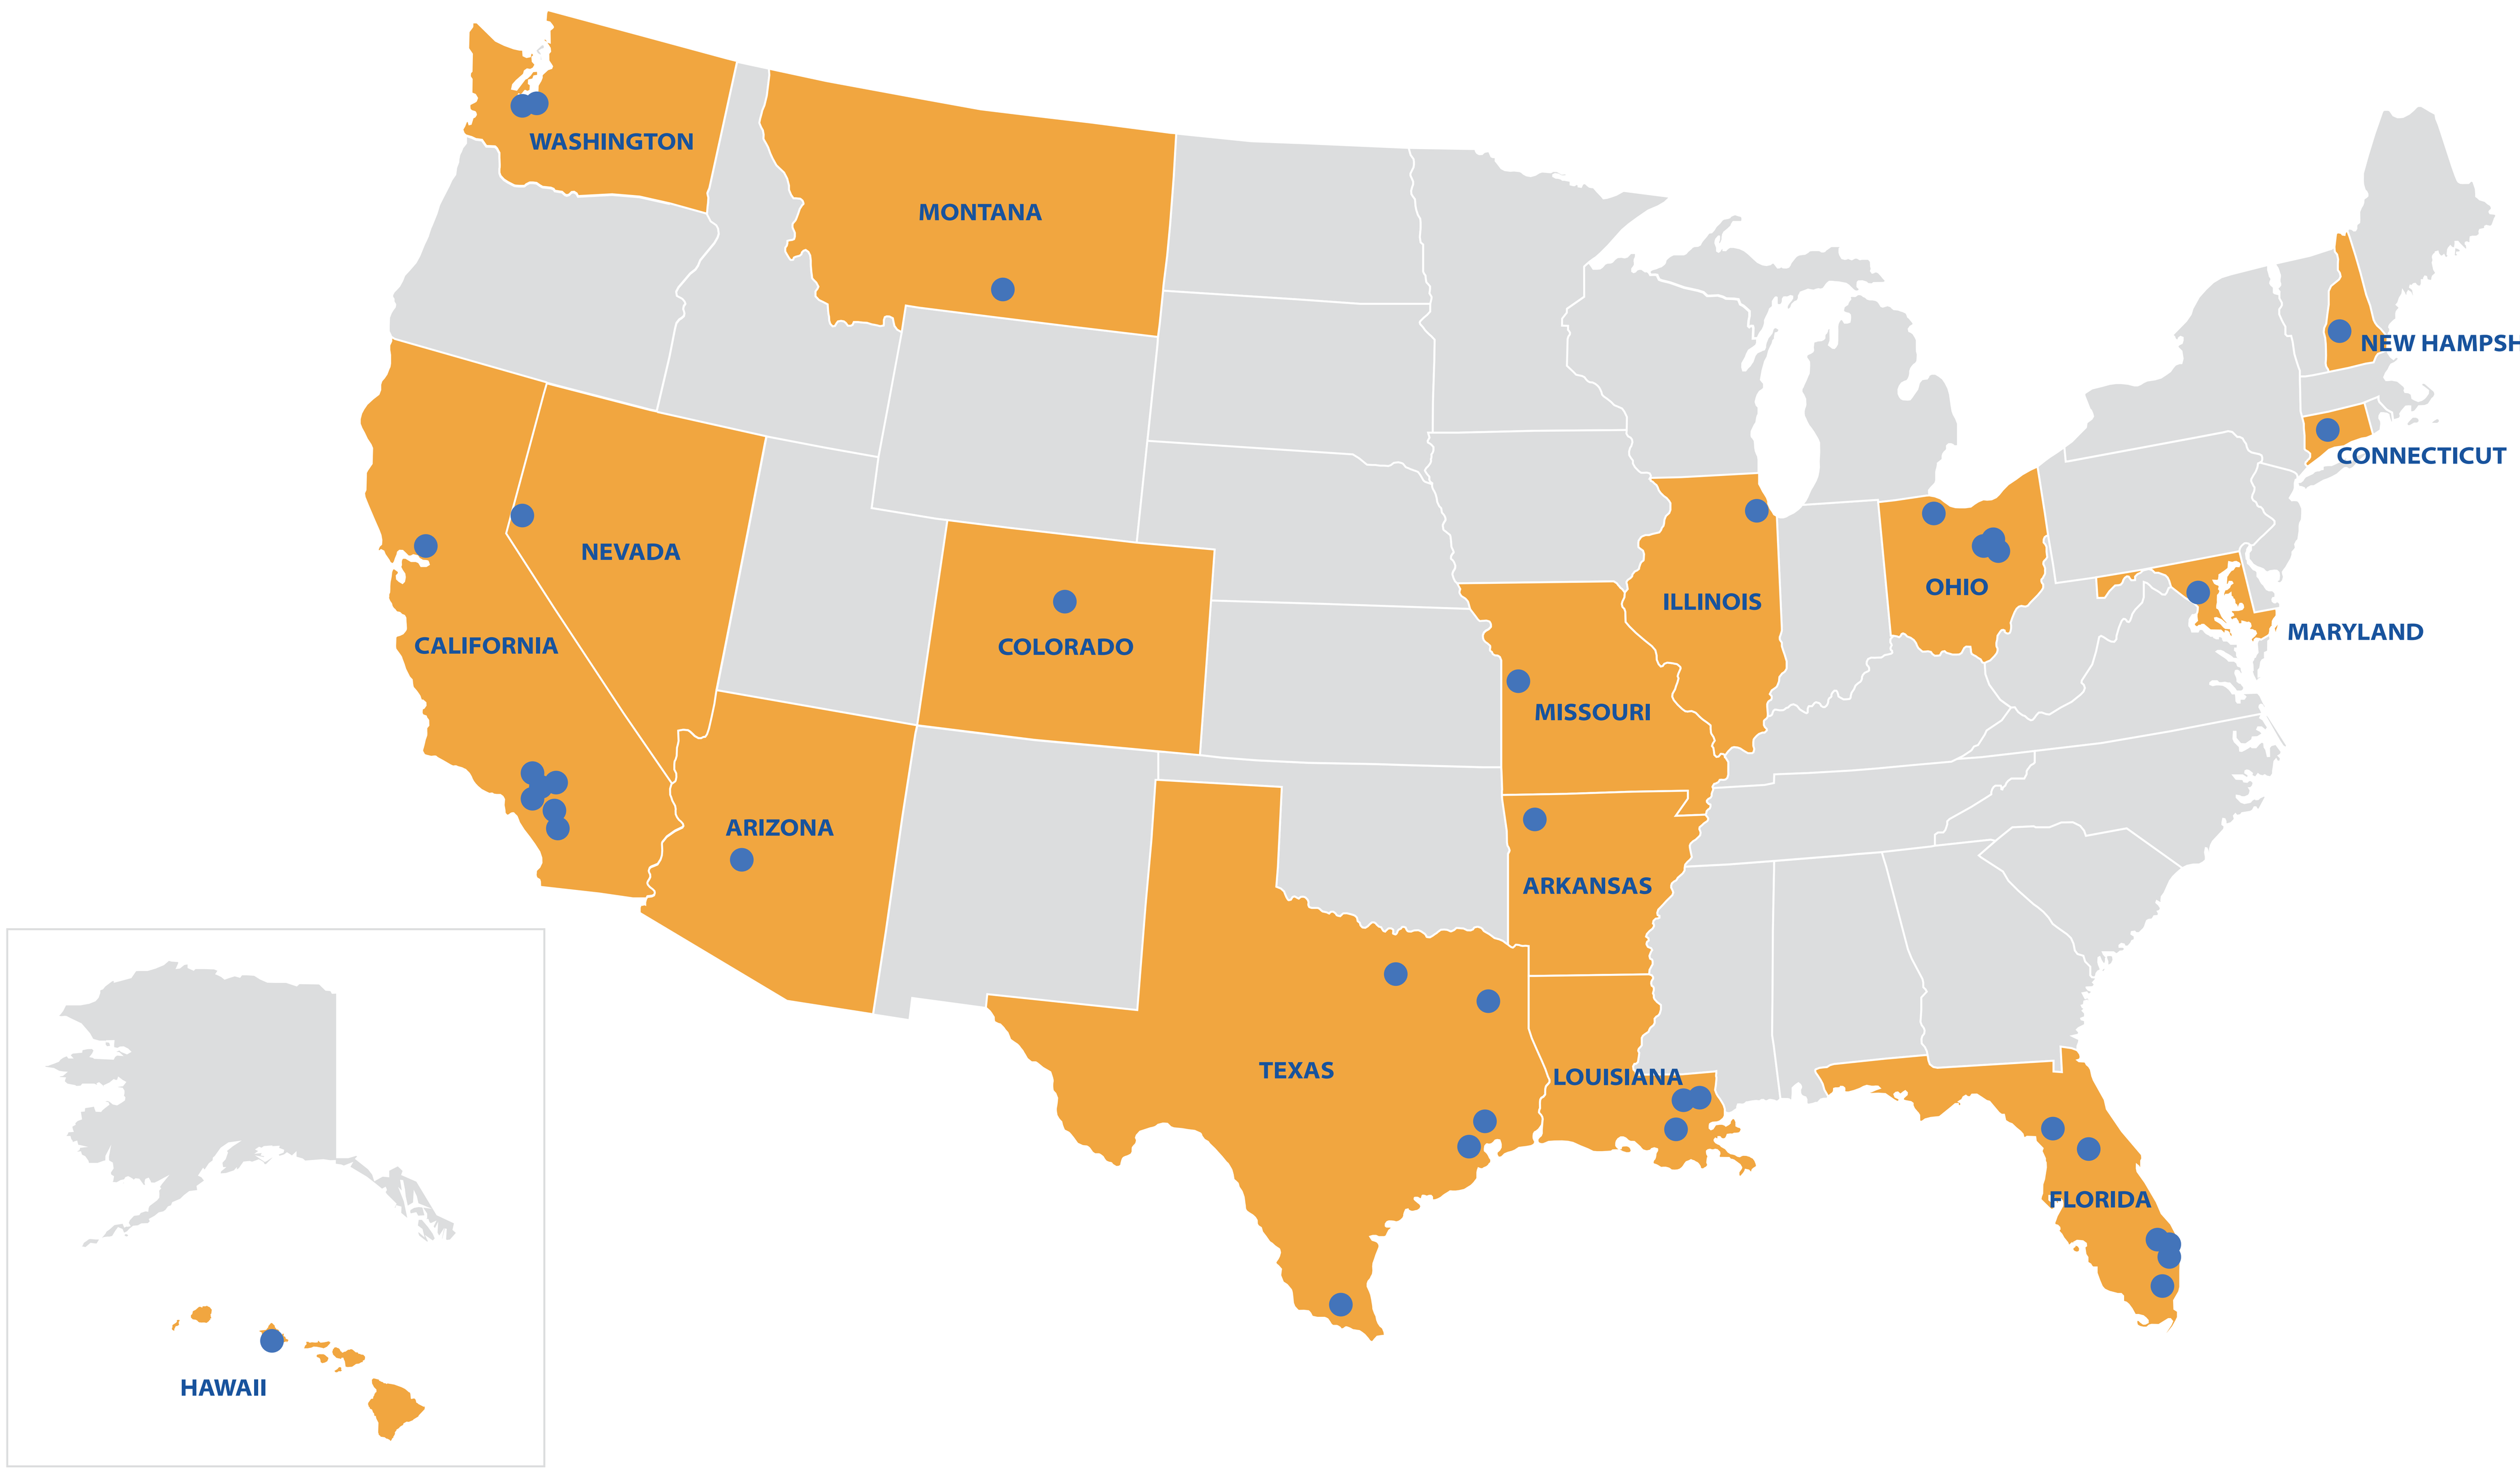
WASHINGTON
MONTANA
NEW HAMPSHIRE
CONNECTICUT
CALIFORNIA
NEVADA
COLORADO
ILLINOIS
OHIO
MARYLAND
MISSOURI
ARIZONA
ARKANSAS
TEXAS
LOUISIANA
FLORIDA
HAWAII

More information about this study can be found at https://clinicaltrials.gov/study/NCT05468489.

Key inclusion criteria

- 18 years and older
- Histologically or cytologically diagnosed with ES-SCLC
- ≥1 measurable lesion
- Stable and treated brain metastases
- ECOG PS 0 or 1
- No significant organ dysfunction
- Expected survival ≥12 weeks

Key exclusion criteria

- Histologically or cytologically confirmed mixed-stage SCLC
- Prior systemic SCLC treatments
- Grade ≥2 peripheral neuropathy
- Ejection fraction <50% or NYHA class III to IV cardiac insufficiency
- Pregnant or breastfeeding females

Support and contact

- This study is sponsored by Shanghai Henlius Biotechnology Co., Ltd
- Correspondence: Marshika Vickers
 - Phone number: (510) 445-0305
 - Email: marshika_vickers@henlius.com

Acknowledgements

- Thank you to the patients and their families who make this trial possible
- Thank you to the clinical study teams and investigators for their work and contributions
- This study is supported by Shanghai Henlius Biotech, Inc. and Fosun Pharma
- All authors contributed to and approved this presentation; writing and editorial assistance were provided by Christine Schwall-Pecci of BGB Group, funded by Fosun Pharma

FPO
Copies of this poster can be obtained through Quick Response (QR). Copies are for personal use only and may not be reproduced without permission of the authors.

Abbreviations

1L, first-line; AE, adverse event; DOR, duration of response; ES-SCLC, extensive stage small cell lung cancer; LS-SCLC, limited stage small cell lung cancer; mAb, monoclonal antibody; ORR, overall response rate; OS, overall survival; PD, pharmacodynamics; PD-1, programmed cell death 1; PD-L1, programmed death-ligand 1; PD-L2, programmed death-ligand 2; PFS, progression-free survival; PK, pharmacokinetics; PS, performance status; Q3W, every 3 weeks; QoL, quality of life; R, randomization; SCLC, small cell lung cancer; SOC, standard of care.

References

1. Rudin CM, Brambilla E, Faivre-Finn C, Sage J. Small-cell lung cancer. *Nat Rev Dis Primer*. 2021;7(1):3. **2.** Oronsky B, Abrouk N, Caroen S, et al. A 2022 update on extensive stage small-cell lung cancer (SCLC). *J Cancer*. 2022;13(9):2945-2953. **3.** Paz-Ares L, Dvorkin M, Chen Y, et al. Durvalumab plus platinum-etoposide versus platinum-etoposide in first-line treatment of extensive-stage small-cell lung cancer (CASPIAN): a randomised, controlled, open-label, phase 3 trial. *Lancet*. 2019;394(10212):1929-1939. **4.** Horn L, Mansfield AS, Szczęsna A, et al. First-line atezolizumab plus chemotherapy in extensive-stage small-cell lung cancer. *N Engl J Med*. 2018;379(23):2220-2229. **5.** Henlius. Research progress. Accessed November 7, 2023. https://www.henlius.com/en/product.html. **6.** Cheng Y, Han L, Wu L, et al. Effect of first-line serplulimab vs placebo added to chemotherapy on survival in patients with extensive-stage small cell lung cancer: The ASTRUM-005 randomized clinical trial. *JAMA*. 2022;328(12):1223-1232. **7.** Henlius. Henlius' novel anti-PD-1 mAb HANSIZHUANG (serplulimab) approved for the treatment of ES-SCLC [press release]. January 17, 2023. Accessed October 11, 2023. https://www.henlius.com/en/NewsDetails-3949-26.html. **8.** Cheng Y. ASTRUM-005: Updated results of first-line serplulimab versus placebo combined with chemotherapy in extensive-stage small-cell lung cancer. Presented at: European Society of Medical Oncology Asia; December 2, 2022; Singapore. **9.** Fosun. Fosun Pharma and Henlius entered into an exclusive license agreement for serplulimab in the US [press release]. January 7, 2023. Accessed November 7, 2023. https://fosunpharmausa.com/fosun-pharma-and-henlius-entered-into-an-exclusive-license-agreement-for-serplulimab-in-the-us/. **10.** Clinicaltrials.gov. NCT05468489. Accessed October 10, 2023. **11.** Issafras H, Fan S, Tseng C, et al. Structural basis of HLX10 PD-1 receptor recognition, a promising anti-PD-1 antibody clinical candidate for cancer immunotherapy. *PLoS One*. 2021;16(12):e0257972. **12.** Fan SL, Jiang WD. Structural basis of HLX10 PD-1 receptor recognition, a promising anti-PD-1 antibody clinical candidate for cancer immunotherapy (2021) https://doi.org/10.2210/pdb7E9B/pdb.